Efficacy and Safety of 144 Weeks of Bulevirtide 2 mg or 10 mg Monotherapy From the Ongoing Phase 3 Study MYR301

LBP-029

Pietro Lampertico[1,2], Soo Aleman[3], Maurizia Brunetto[4,5], Antje Blank[6], Pietro Andreone[7], Pavel Bogomolov[8], Vladimir Chulanov[9], Nina Mamonova[10], Natalia Geyvandova[11], Viacheslav Morozov[12], Olga Sagalova[13], Tatyana Stepanova[14], Grace M Chee[15], Dmitry Manuilov[15], Mingyang Li[15], Steve Tseng[15], Audrey H Lau[15], Anu Osinusi[15], Julian Schulze zur Wiesch[16], Markus Cornberg[17], Stefan Zeuzem[18], Heiner Wedemeyer[17]

[1]Division of Gastroenterology and Hepatology, Foundation IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [2]Department of Pathophysiology and Transplantation, CRC "A. M. and A. Migliavacca" Center for Liver Disease, University of Milan, Milan, Italy; [3]Department of Infectious Diseases, Karolinska University Hospital/Karolinska Institutet, Stockholm, Sweden; [4]Hepatology Unit, Reference Center of the Tuscany Region for Chronic Liver Disease and Cancer, University Hospital of Pisa, Pisa, Italy; [5]Department of Clinical and Experimental Medicine, University of Pisa, Pisa, Italy; [6]Medical Faculty Heidelberg/Heidelberg University Hospital, Department of Clinical Pharmacology and Pharmacoepidemiology, Heidelberg University Hospital, Heidelberg, Germany; [7]Internal Medicine, Baggiovara Hospital, University of Modena and Reggio Emilia, Modena, Italy; [8]M.F. Vladimirsky Moscow Regional Research and Clinical Institute, Moscow, Russian Federation; [9]Sechenov University, Moscow, Russian Federation; [10]FSBI National Research Medical Center for Phthisiopulmonology and Infectious Diseases of the Ministry of Health of the Russian Federation, Moscow, Russian Federation; [11]Stavropol Regional Hospital, Stavropol, Russian Federation; [12]LLC Medical Company "Hepatolog," Samara, Russian Federation; [13]South Ural State Medical University, Chelyabinsk, Russian Federation; [14]LLC Clinic of Modern Medicine, Moscow, Russian Federation; [15]Gilead Sciences, Inc., Foster City, CA, USA; [16]Hepatology Outpatient Medical Clinic, University Hospital Hamburg-Eppendorf, Hamburg, Germany; [17]Clinic for Gastroenterology, Hepatology, Infectious Diseases, and Endocrinology, Hannover Medical School, Hannover, Germany; [18]Department of Medicine, University Hospital Frankfurt, Frankfurt, Germany

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- Long-term treatment with BLV monotherapy over 144 weeks remained safe and effective
- Improvements in virologic and biochemical responses and liver stiffness, as well as low occurrence of liver-related outcomes, are supportive of the potential clinical benefits of long-term BLV monotherapy

Plain Language Summary

- Patients with chronic hepatitis delta who received bulevirtide for 144 weeks achieved substantial reductions in their hepatitis delta virus RNA levels and had either improved or maintained key markers of liver health and function

References: **1.** Stockdale AJ, et al. *J Hepatol*. 2020;73:523-32. **2.** Alfaiate D, et al. *J Hepatol*. 2020;73(3):533-9. **3.** Rizzetto M, et al. *J Hepatol*. 2021;74(5):1200-11. **4.** Hepcludex. European Medicines Agency SmPC. Gilead Sciences, Inc.; 2023. **5.** European Association for the Study of the Liver. *J Hepatol*. 2023;79:433-60. **6.** Wedemeyer H, et al. *J Hepatol*. Published online May 9, 2024.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators and their corresponding site staff. Editing and production assistance were provided by Molly Yeager, PhD, of AlphaScientia, a Red Nucleus company, and were funded by Gilead Sciences, Inc. This study was funded by Gilead Sciences, Inc.

Disclosures: **PL** reports speaking and teaching fees from and participation in advisory committees or review panels for AbbVie; Aligos Therapeutics; Alnylam Pharmaceuticals; Antios Therapeutics; Arrowhead Pharmaceuticals; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; GSK; Janssen; Merck Sharp & Dohme; MYR GmbH; Roche; and Spring Bank Pharmaceuticals. **SA** reports speaking honoraria from AbbVie; Biogen; Gilead Sciences, Inc.; and Merck Sharp & Dohme, and research grants from AbbVie and Gilead Sciences, Inc. **MB** reports consulting, speaker fees, and research support from AbbVie; Eisai-MSD; Gilead Sciences, Inc.; Janssen; and Roche. **AB** reports research funding from MYR GmbH. **PA** reports being a speaker, consultant, and advisory board member for AbbVie, Boehringer Ingelheim, Bristol Myers Squibb, and Gilead Sciences, Inc. **PB** reports nothing to disclose. **VC** reports being a consultant and giving sponsored lectures for AbbVie; AstraZeneca; Bristol Myers Squibb; Gilead Sciences, Inc.; GSK; Hepatera; Merck Sharp & Dohme; Roche; and R-Pharm. **NM**, **NG**, **VM**, **OS**, and **TS** report nothing to disclose. **GMC**, **DM**, **ML**, **ST**, **AHL**, and **AO** are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc. **JSW** reports consultation and lecture fees for Gilead Sciences, Inc. **MC** reports honoraria from AbbVie; Falk; Gilead Sciences, Inc.; GSK; Janssen-Cilag; Merck Sharp & Dohme; Novartis; Roche; Spring Bank Pharmaceuticals; and Swedish Orphan Biovitrum. **SZ** reports speakers bureau and/or consultancy for AbbVie; Allergan; BioMarin; Gilead Sciences, Inc.; Intercept Pharmaceuticals; Janssen; Merck Sharp & Dohme; Novo Nordisk; Swedish Orphan Biovitrum; and Theratechnologies. **HW** reports honoraria for speaking or consulting from Abbott; AbbVie; Boehringer Ingelheim; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; Janssen; Merck Sharp & Dohme; MYR GmbH; Novartis; Novira; Roche; Roche Diagnostics; Siemens; and Transgene; and research support from Abbott, Bristol Myers Squibb, and Gilead Sciences, Inc.

Introduction

- Hepatitis delta virus (HDV) infection affects between 10 and 20 million people worldwide[1] and causes the most severe form of chronic viral hepatitis[2,3]
- Bulevirtide (BLV) is a first-in-class entry inhibitor approved in the EU, Great Britain, Switzerland, and the Russian Federation for treatment of chronic hepatitis delta (CHD)[4,5] and is recommended by the European Association for the Study of the Liver (EASL) guidelines for the treatment of CHD in patients with compensated liver disease[5]
- Previously published results demonstrated that BLV 2 mg or 10 mg monotherapy for 96 weeks leads to HDV RNA reductions and alanine aminotransferase (ALT) improvements, and is generally safe[6]

Objective

- The objective of this interim analysis was to evaluate the long-term efficacy and safety of BLV 2 mg or 10 mg monotherapy through 144 weeks of therapy

Methods

MYR301 Study Design

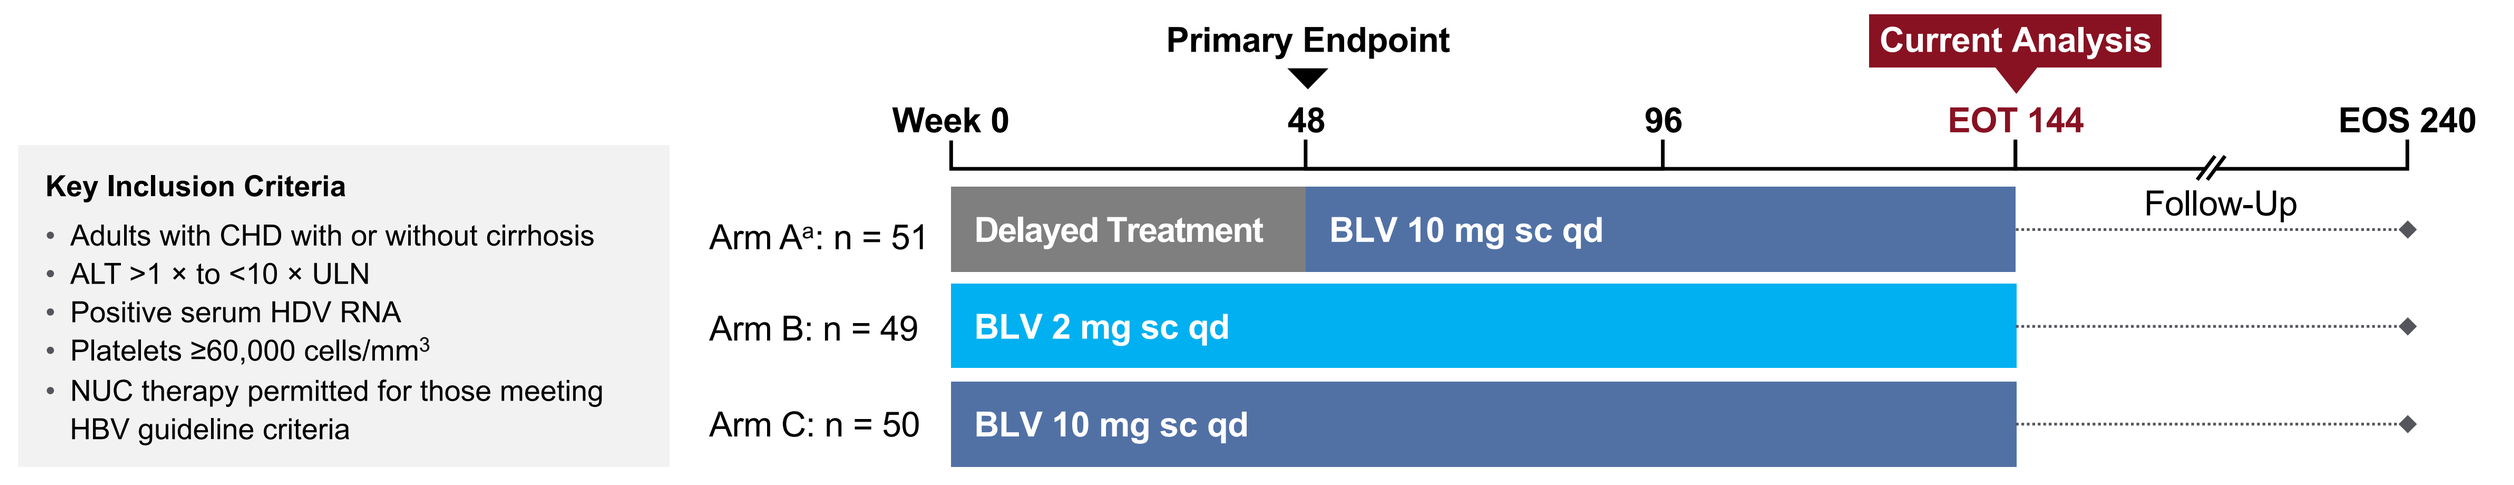
Primary Endpoint
Current Analysis
Week 0
48
96
EOT 144
EOS 240
Follow-Up
Key Inclusion Criteria
Adults with CHD with or without cirrhosis
ALT >1 × to <10 × ULN
Positive serum HDV RNA
Platelets ≥60,000 cells/mm³
NUC therapy permitted for those meeting HBV guideline criteria
Arm A[a]: n = 51
Delayed Treatment
BLV 10 mg sc qd
Arm B: n = 49
BLV 2 mg sc qd
Arm C: n = 50
BLV 10 mg sc qd

[a]One patient discontinued the study prior to receiving BLV; therefore, n = 50 was used for all arm A efficacy results, with baseline reset at the week 48 visit, and efficacy presented by BLV duration (not including the delayed treatment period).
BLV, bulevirtide; EOS, end of study; EOT, end of treatment; NUC, nucleos(t)ide analogue; qd, once daily; sc, subcutaneous.

- The multicentre, open-label, randomised, Phase 3 study MYR301 (NCT03852719) was conducted at 16 sites across 4 countries (Germany, Italy, Russian Federation, and Sweden)
- Primary endpoint: the proportion of patients achieving a combined response (undetectable HDV RNA or ≥2 $\log_{10}$ IU/mL decline from baseline [BL] and ALT normalisation) at week (W) 48
 — HDV RNA was manually extracted and viral load was quantified using RoboGene 2.0® (lower limit of quantification, 50 IU/mL)
- Endpoints evaluated over 144 weeks: the proportion of patients achieving key efficacy endpoints (virologic and biochemical, change in liver stiffness, liver-related outcomes, changes in liver chemistries, and adverse events (AEs)
- Univariate logistic regression was conducted to evaluate if BL characteristics predicted undetectable HDV RNA at W144 for patients in arms B and C who completed W144 and had BL HDV RNA ≥250 IU/mL; predictors with p-value <.05 were considered significant

Results

Baseline Demographics and Disease Characteristics

	BLV 2 mg (n = 49)	BLV 10 mg (n = 50)	Delayed Treatment to BLV 10 mg (n = 51)
Age, years, mean (SD)	44 (9)	41 (9)	41 (8)
Male sex, n (%)	30 (61)	30 (60)	26 (51)
Race,[a] n (%)			
White	41 (84)	43 (86)	40 (78)
Asian	8 (16)	6 (12)	11 (22)
Cirrhosis present, n (%)	23 (47)	24 (48)	24 (47)
Liver stiffness, kPa, mean (SD)	14.0 (8.2)	14.8 (9.3)	15.3 (9.0)
ALT, U/L, mean (SD)	108 (63)	123 (81)	102 (62)
HDV RNA, $\log_{10}$ IU/mL, mean (SD)	5.10 (1.20)	4.96 (1.46)	5.08 (1.36)
Genotype HDV-1,[b] n (%)	49 (100)	48 (96)	51 (100)
HBsAg, $\log_{10}$ IU/mL, mean (SD)	3.67 (0.52)	3.61 (0.59)	3.68 (0.47)
HBV DNA, $\log_{10}$ IU/mL, mean (SD)	1.30 (1.29)	1.08 (1.26)	0.89 (0.99)
HBV genotype, n (%)			
A	2 (4)	2 (4)	2 (4)
D	47 (96)	44 (88)	44 (86)
Other[c]/missing	0	4 (8)	5 (10)
Previous IFN therapy, n (%)	26 (53)	29 (58)	29 (57)
Concomitant HBV NUC treatment,[d] n (%)	32 (65)	27 (54)	32 (63)

[a]BLV 10 mg arm: Black, n = 1. [b]BLV 10 mg arm: HDV GT 5, n = 1; missing HDV GT, n = 1. [c]BLV 10 mg arm: HBV GT E, n = 1; delayed treatment to BLV 10 mg arm: unclassified HBV GT, n = 2. [d]All patients who started NUC therapy at or post BL, except 1, started at BL or within 2 days of BL.
ALT, alanine aminotransferase; BL, baseline; BLV, bulevirtide; GT, genotype; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; HDV, hepatitis delta virus; IFN, interferon; NUC, nucleos(t)ide analogue; W, week.

- Patient retention remained high through 144 weeks with no discontinuations related to study treatment; study completion rates are below:
 — Arm A: 96% (49/51); pregnancy, 1; death (plasma cell myeloma unrelated to BLV), 1
 — Arm B: 92% (45/49); withdrew consent, 3; pregnancy, 1
 — Arm C: 88% (44/50); withdrew consent, 5; physician decision, 1

Key Efficacy Over Time (by BLV Duration)

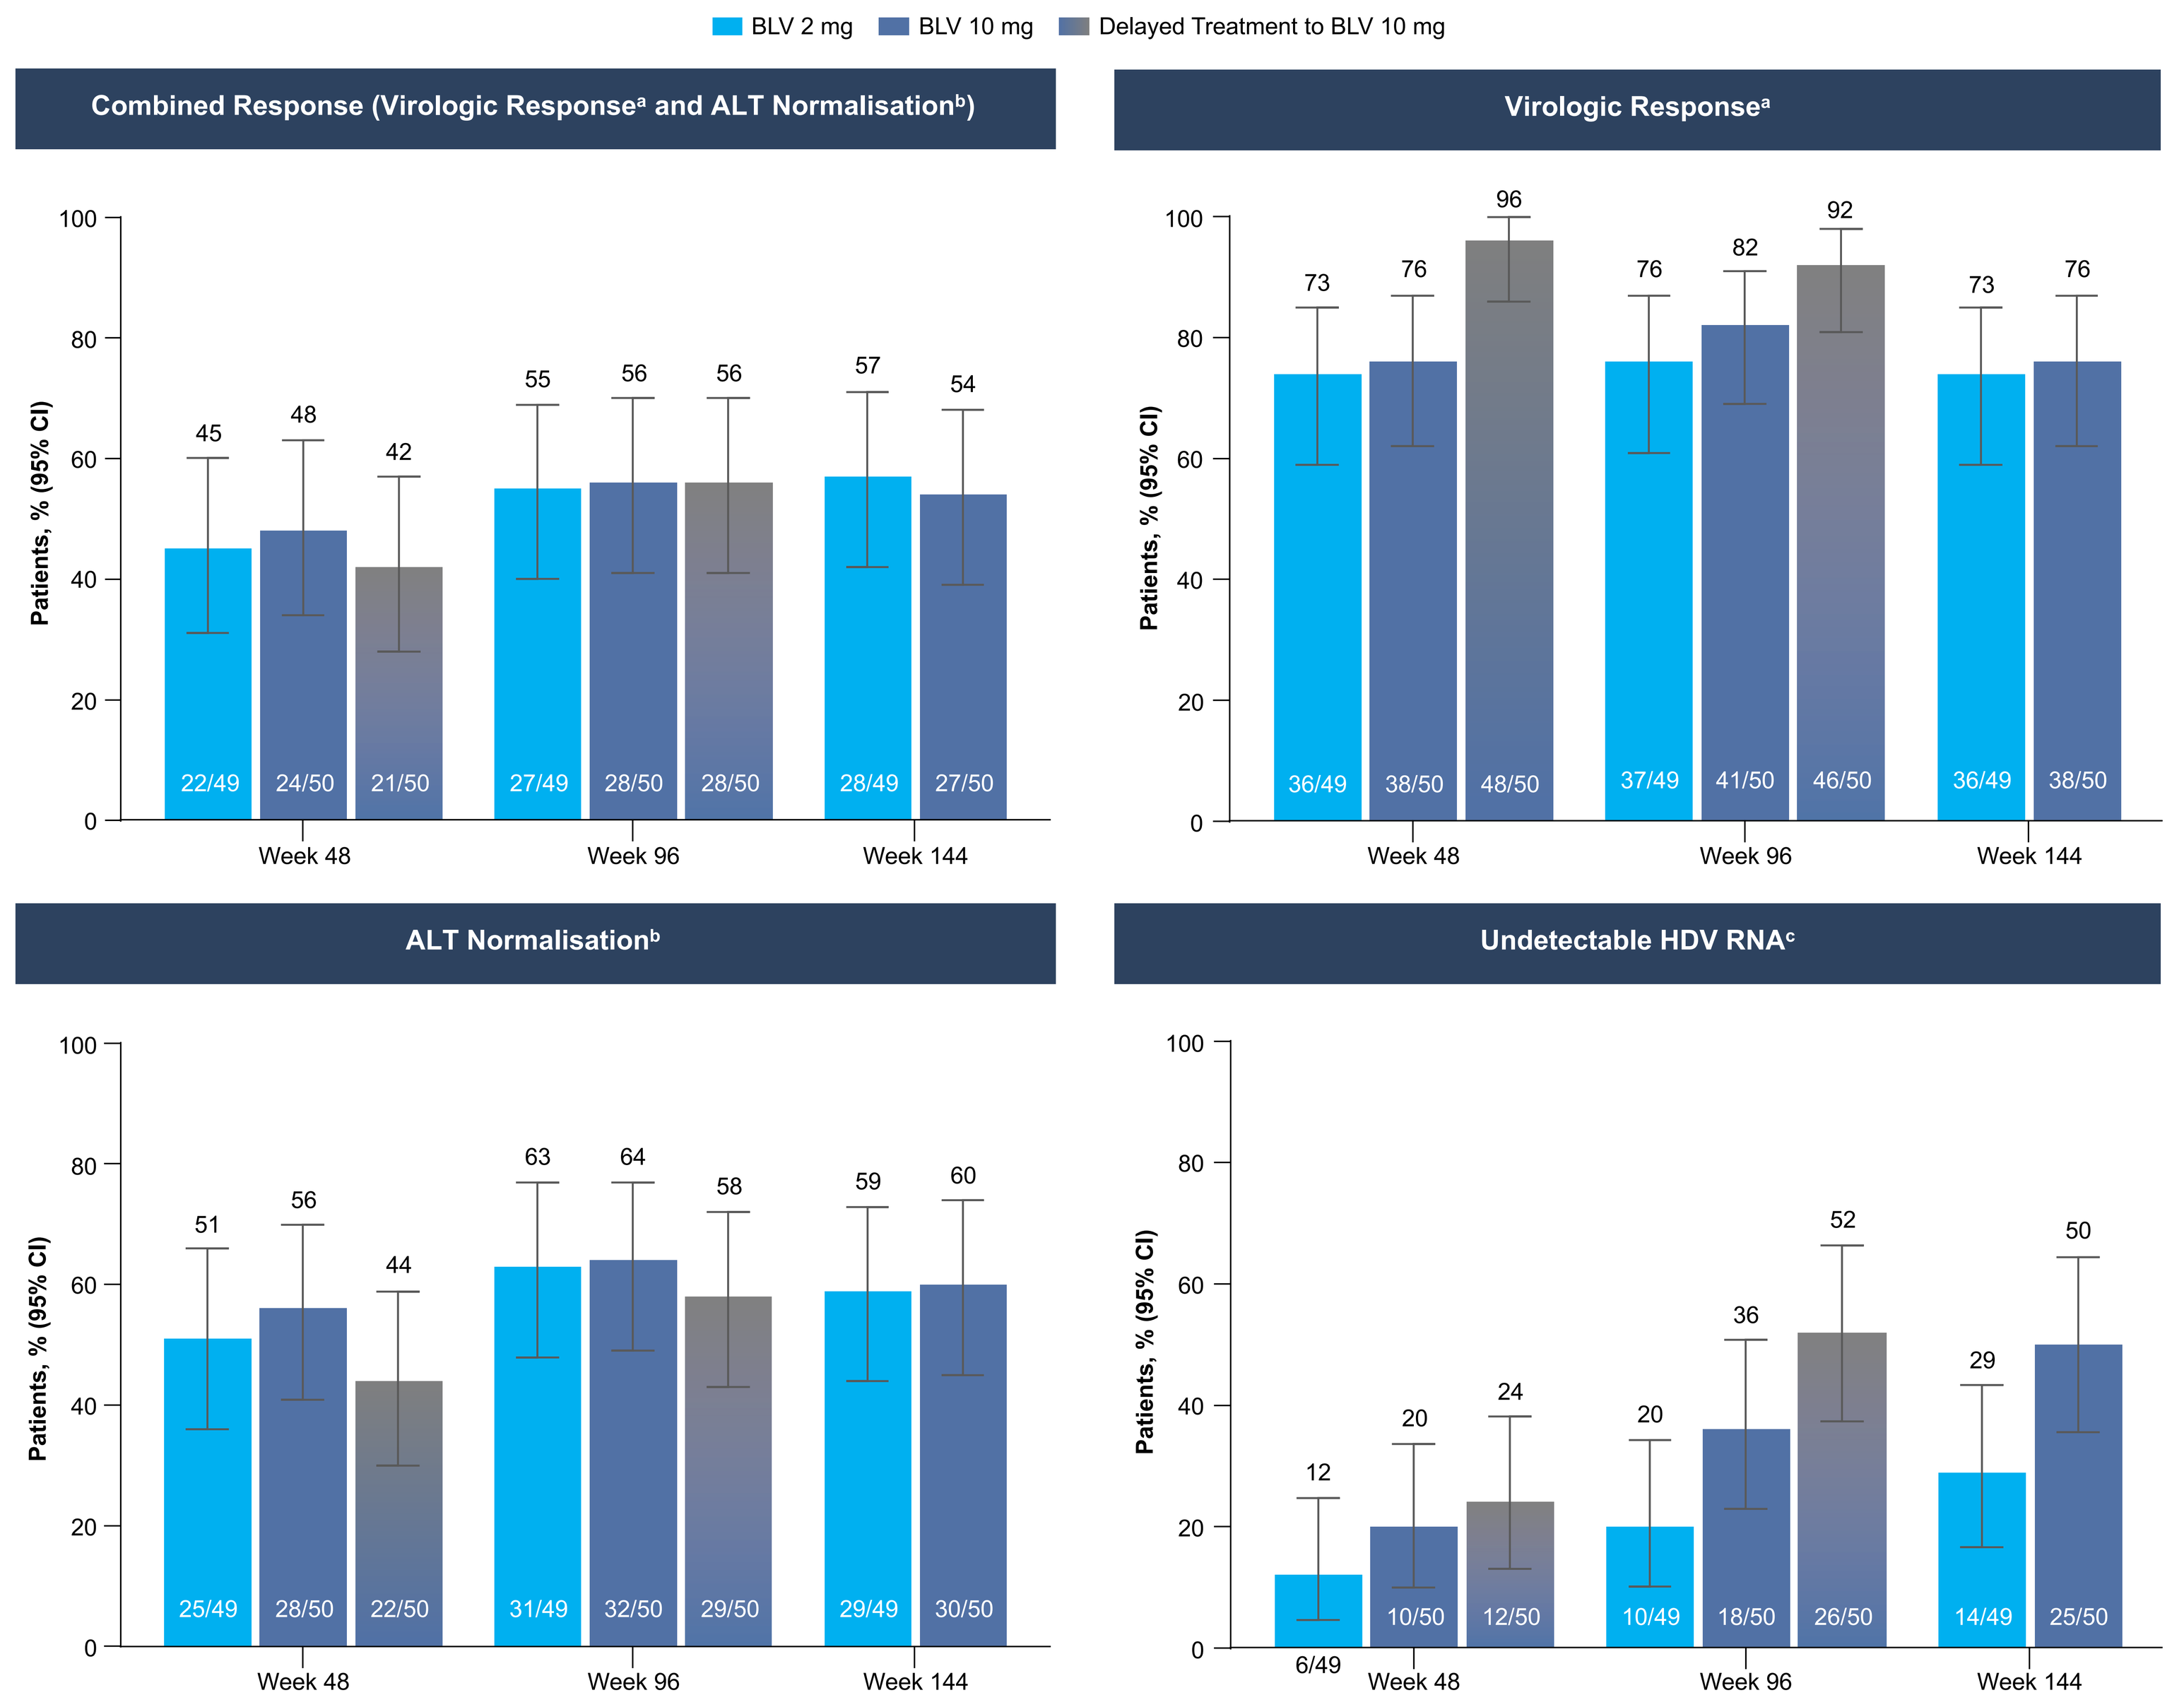
BLV 2 mg
BLV 10 mg
Delayed Treatment to BLV 10 mg
Combined Response (Virologic Response[a] and ALT Normalisation[b])
Virologic Response[a]
ALT Normalisation[b]
Undetectable HDV RNA[c]
Patients, % (95% CI)
45 48 42 55 56 56 57 54
22/49 24/50 21/50 27/49 28/50 28/50 28/49 27/50
73 76 96 76 82 92 73 76
36/49 38/50 48/50 37/49 41/50 46/50 36/49 38/50
51 56 44 63 64 58 59 60
25/49 28/50 22/50 31/49 32/50 29/50 29/49 30/50
12 20 24 20 36 52 29 50
6/49 10/50 12/50 10/49 18/50 26/50 14/49 25/50
Week 48
Week 96
Week 144

Patients with missing values were considered nonresponders. [a]Undetectable HDV RNA or ≥2 $\log_{10}$ IU/mL decline from BL. [b]ALT normalisation was defined at Russian sites as ≤31 U/L for females and ≤41 U/L for males, and at all other sites as ≤34 U/L for females and ≤49 U/L for males. [c]Undetectable HDV RNA was defined as <LLOQ (50 IU/mL; target not detected).
ALT, alanine aminotransferase; BL, baseline; BLV, bulevirtide; HDV hepatitis delta virus; LLOQ, lower limit of quantification.

- Combined response, ALT normalisation, and virologic response rates increased through W96 and were maintained from W96 to W144; the response rates were similar between the BLV 2 mg and 10 mg arms
- Rates of undetectable HDV RNA continually increased over 144 weeks for both the BLV 2 mg and 10 mg arms; rates were numerically higher for the BLV 10 mg arm
- Among patients who achieved undetectable HDV RNA at any post-BL time points, those in the BLV 10 mg arm did so faster than in the BLV 2 mg arm
 — The mean (SD) number of weeks after starting BLV treatment to first reaching undetectable HDV RNA was as follows:
 ○ BLV 2 mg: 77 (45)
 ○ BLV 10 mg: 69 (41)

Baseline Predictors of Undetectable HDV RNA at W144

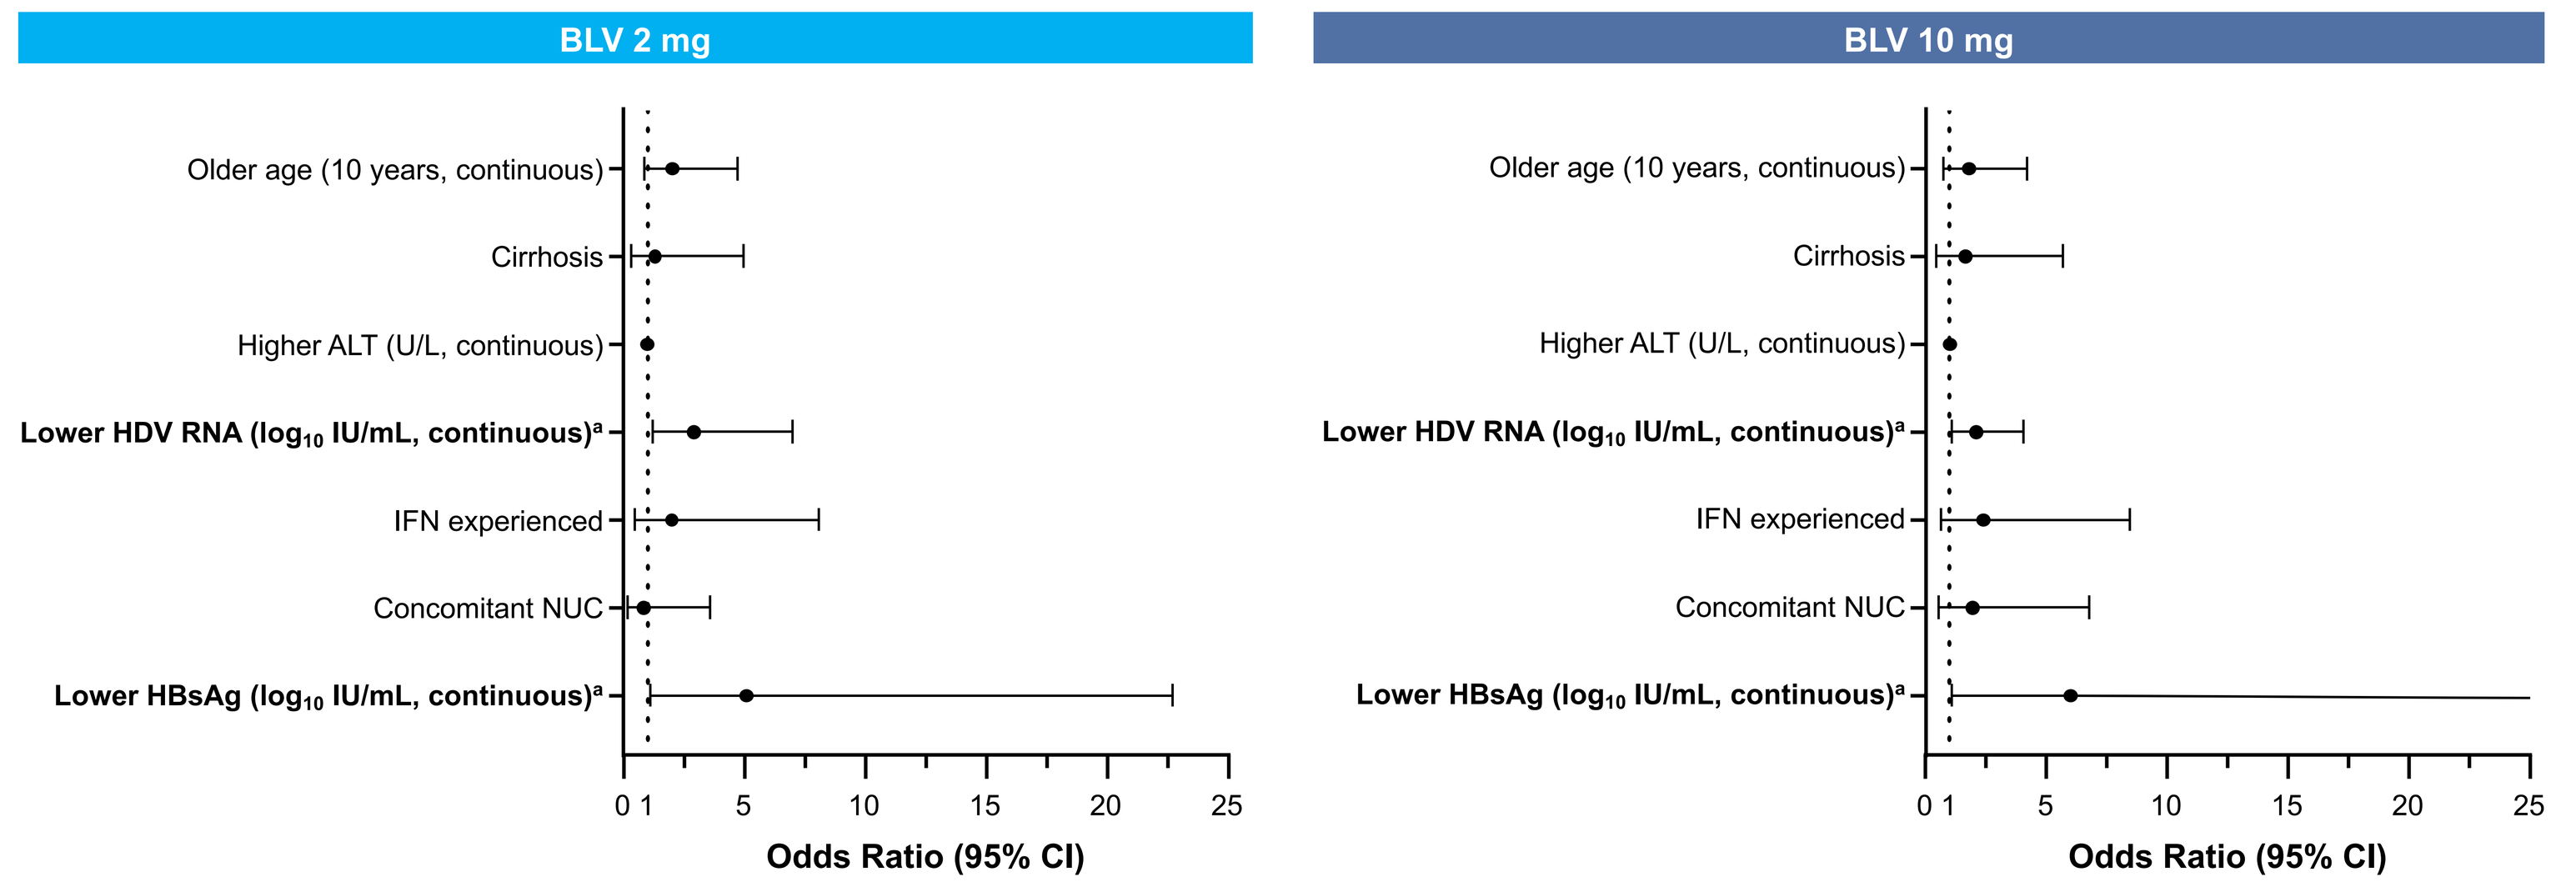
BLV 2 mg
BLV 10 mg
Older age (10 years, continuous)
Cirrhosis
Higher ALT (U/L, continuous)
Lower HDV RNA ($\log_{10}$ IU/mL, continuous)[a]
IFN experienced
Concomitant NUC
Lower HBsAg ($\log_{10}$ IU/mL, continuous)[a]
0 1 5 10 15 20 25
Odds Ratio (95% CI)

[a]p <0.05. The upper CI boundary for lower HBsAg with BLV 10 mg (32) is not shown due to the figure scale. The analysis population included patients who completed W144 with BL HDV RNA ≥250 IU/mL.
ALT, alanine aminotransferase; BL, baseline; BLV, bulevirtide; HBsAg, hepatitis B surface antigen; HDV, hepatitis delta virus; IFN, interferon; NUC, nucleos(t)ide analogue; W, week.

- Lower HDV RNA and hepatitis B surface antigen (HBsAg) levels at BL were identified as potential predictors of undetectable HDV RNA at W144

Platelet Count, Liver Chemistries, and Other Efficacy Markers Over Time

	BLV 2 mg (n = 49)			BLV 10 mg (n = 50)		
	BL	Week 96	Week 144	BL	Week 96	Week 144
Platelets, × 10^9/L, mean (SD)	153 (52.5)	169 (69.1)	162 (62.5)	160 (53.1)	174 (70.7)	163 (63.5)
Platelets <90 × 10^9/L, %	14	13	14	12	11	14
Total bilirubin, µmol/L, median (Q1, Q3)	10.4 (6.7, 14.7)	12.7 (7.5, 15.9)	13.6 (8.6, 19.0)	10.5 (7.6, 13.6)	10.2 (7.5, 14.5)	11.8 (7.2, 15.0)
Albumin, g/L, mean (SD)	44 (3.1)	45 (2.6)	47 (2.9)	44 (3.2)	45 (3.2)	46 (3.3)
Albumin <35 g/L, %	0	0	0	2	2	0
GGT, U/L, median (Q1, Q3)	57 (27, 73)	27 (18, 37)	25 (18, 37)	42 (27, 73)	24 (17, 36)	23 (19, 39)
INR, mean (SD)	1.12 (0.111)	1.10 (0.116)	1.12 (0.145)	1.14 (0.112)	1.10 (0.129)	1.14 (0.222)
HBsAg, $\log_{10}$ IU/mL, mean (SD)	3.67 (0.515)	3.47 (0.578)	3.29 (0.866)	3.61 (0.593)	3.47 (0.638)	3.41 (0.583)
Liver Stiffness, kPa, mean (SD)	13.99 (8.190)	10.2 (6.434)	9.43 (5.281)	14.81 (9.265)	10.04 (6.754)	11.28 (9.836)
Liver stiffness >15 kPa, %	31	15	11	30	15	19

Values are based on available data.
BL, baseline; BLV, bulevirtide; GGT, gamma-glutamyl transferase; HBsAg, hepatitis B surface antigen; INR, international normalised ratio; LS, least square; n/a, not applicable; Q, quartile.

- Platelet counts, liver chemistries, liver stiffness, and HBsAg values remained stable or improved over 144 weeks of BLV monotherapy, including for the subgroup with cirrhosis (data not shown)

Liver-Related Outcomes and Other Endpoints

- No patients had progression to liver-related outcomes over 144 weeks, except for 1 case of mild ascites (arm A) that occurred 165 days after switching to BLV 10 mg in a patient with cirrhosis at BL; the ascites resolved while on BLV
- Only 1 patient experienced HBsAg loss in the delayed treatment to BLV 10 mg arm, and no participant experienced HBsAg seroconversion

Overall Safety Summary

	BLV 2 mg (n = 49)		BLV 10 mg (n = 50)		Delayed Treatment to BLV 10 mg (n = 50)	
Patients, n (%)	Week 96	Week 144	Week 96	Week 144	Week 48–Week 96[a]	Week 48–Week 144
Any AE	47 (96)	48 (98)	48 (96)	48 (96)	42 (84)	46 (92)
Any grade 3–4 AE	9 (18)	12 (24)	8 (16)	10 (20)	3 (6)	5 (10)
Any SAE	2 (4)	3 (6)	4 (8)	6 (12)	2 (4)	3 (6)
Any SAE related to BLV	0	0	0	0	0	0
Any AE leading to withdrawal of BLV	0	0	0	0	0	0
Any AE related to BLV	25 (51)	27 (55)	36 (72)	37 (74)	22 (44)	23 (46)
Death	0	0	0	0	1 (2)[b]	1 (2)[b]
Laboratory abnormalities, total bile salts (µmol/L), mean (SD)	24 (26)	28 (40)	52 (83)	59 (63)	63 (50)[c]	64 (59)[c]
AEs of interest[d]						
Headache	9 (18)	10 (20)	12 (24)	12 (24)	7 (14)	7 (14)
Dizziness	2 (4)	2 (4)	4 (8)	4 (8)	1 (2)	1 (2)
Nausea	3 (6)	3 (6)	6 (12)	6 (12)	1 (2)	1 (2)
Pruritus	6 (12)	6 (12)	9 (18)	8 (16)	0	0
Fatigue	7 (14)	7 (14)	9 (18)	9 (18)	2 (4)	3 (6)
ISR[e]	10 (20)	10 (20)	15 (30)	15 (30)	6 (12)	8 (16)

All AEs were treatment emergent. AEs from the first 96 weeks are from Wedemeyer H, et al. (2024)[6]. [a]AEs from the first 48 weeks are not shown. [b]One death due to plasma cell myeloma not related to study treatment. [c]For the delayed treatment to 10 mg arm, week 96 and 144 total bile salt values reflect week 48 and 96 of BLV 10 mg treatment, respectively. [d]AEs of interest that occurred at higher frequencies in BLV treatment groups compared with the delayed treatment group. [e]Grouped term including injection-site AEs (bruising, induration, reaction, erythema, pruritus, swelling, pain, haematoma, rash, dermatitis).
AE, adverse event; BLV, bulevirtide; ISR, injection-site reaction; SAE, serious adverse event.

- There were no drug discontinuations, serious AEs, or deaths attributed to BLV monotherapy and no reports of HBV reactivation (regardless of concomitant nucleos[t]ide analogue therapy) through 144 weeks of treatment
- The safety profile was similar between the BLV 2 mg and 10 mg arms, except for injection-site reactions, which were more frequent in the BLV 10 mg arm (likely due to 2 daily BLV injections vs 1 daily injection with BLV 2 mg)
- Dose-dependent elevations in bile acid salt remained asymptomatic and were not associated with any clinical sequelae throughout 144 weeks of treatment

European Association for the Study of the Liver; 05–08 June 2024; Milan, Italy